Safe and Dangerous Home Remedies Used for Oral Pain and Discomfort

K Schroeder, RDH, MS; M Santoro, MPH; S Raskin, PhD, MPH; E Tranby, PhD

Introduction

Americans who do not have access to dental care or need to delay their care may resort to home remedies to alleviate mouth pain or discomfort. Having a relationship with an oral health provider might make the difference between choosing safe and effective pain management strategies or choosing risky pain management strategies that may cause additional pain or discomfort.

Methods

- In CareQuest Institute for Oral Health's **2022 State of Oral Health Equity in America (SOHEA) survey**, individuals were asked questions about their oral health and whether they had used home remedies to help alleviate oral health symptoms in the past 12 months.
- Results were collected by NORC at the University of Chicago in January–February 2022 from **adults 18 and older with a final sample size of 5,682**.
- All results presented are statistically significant at the p<0.05 level.

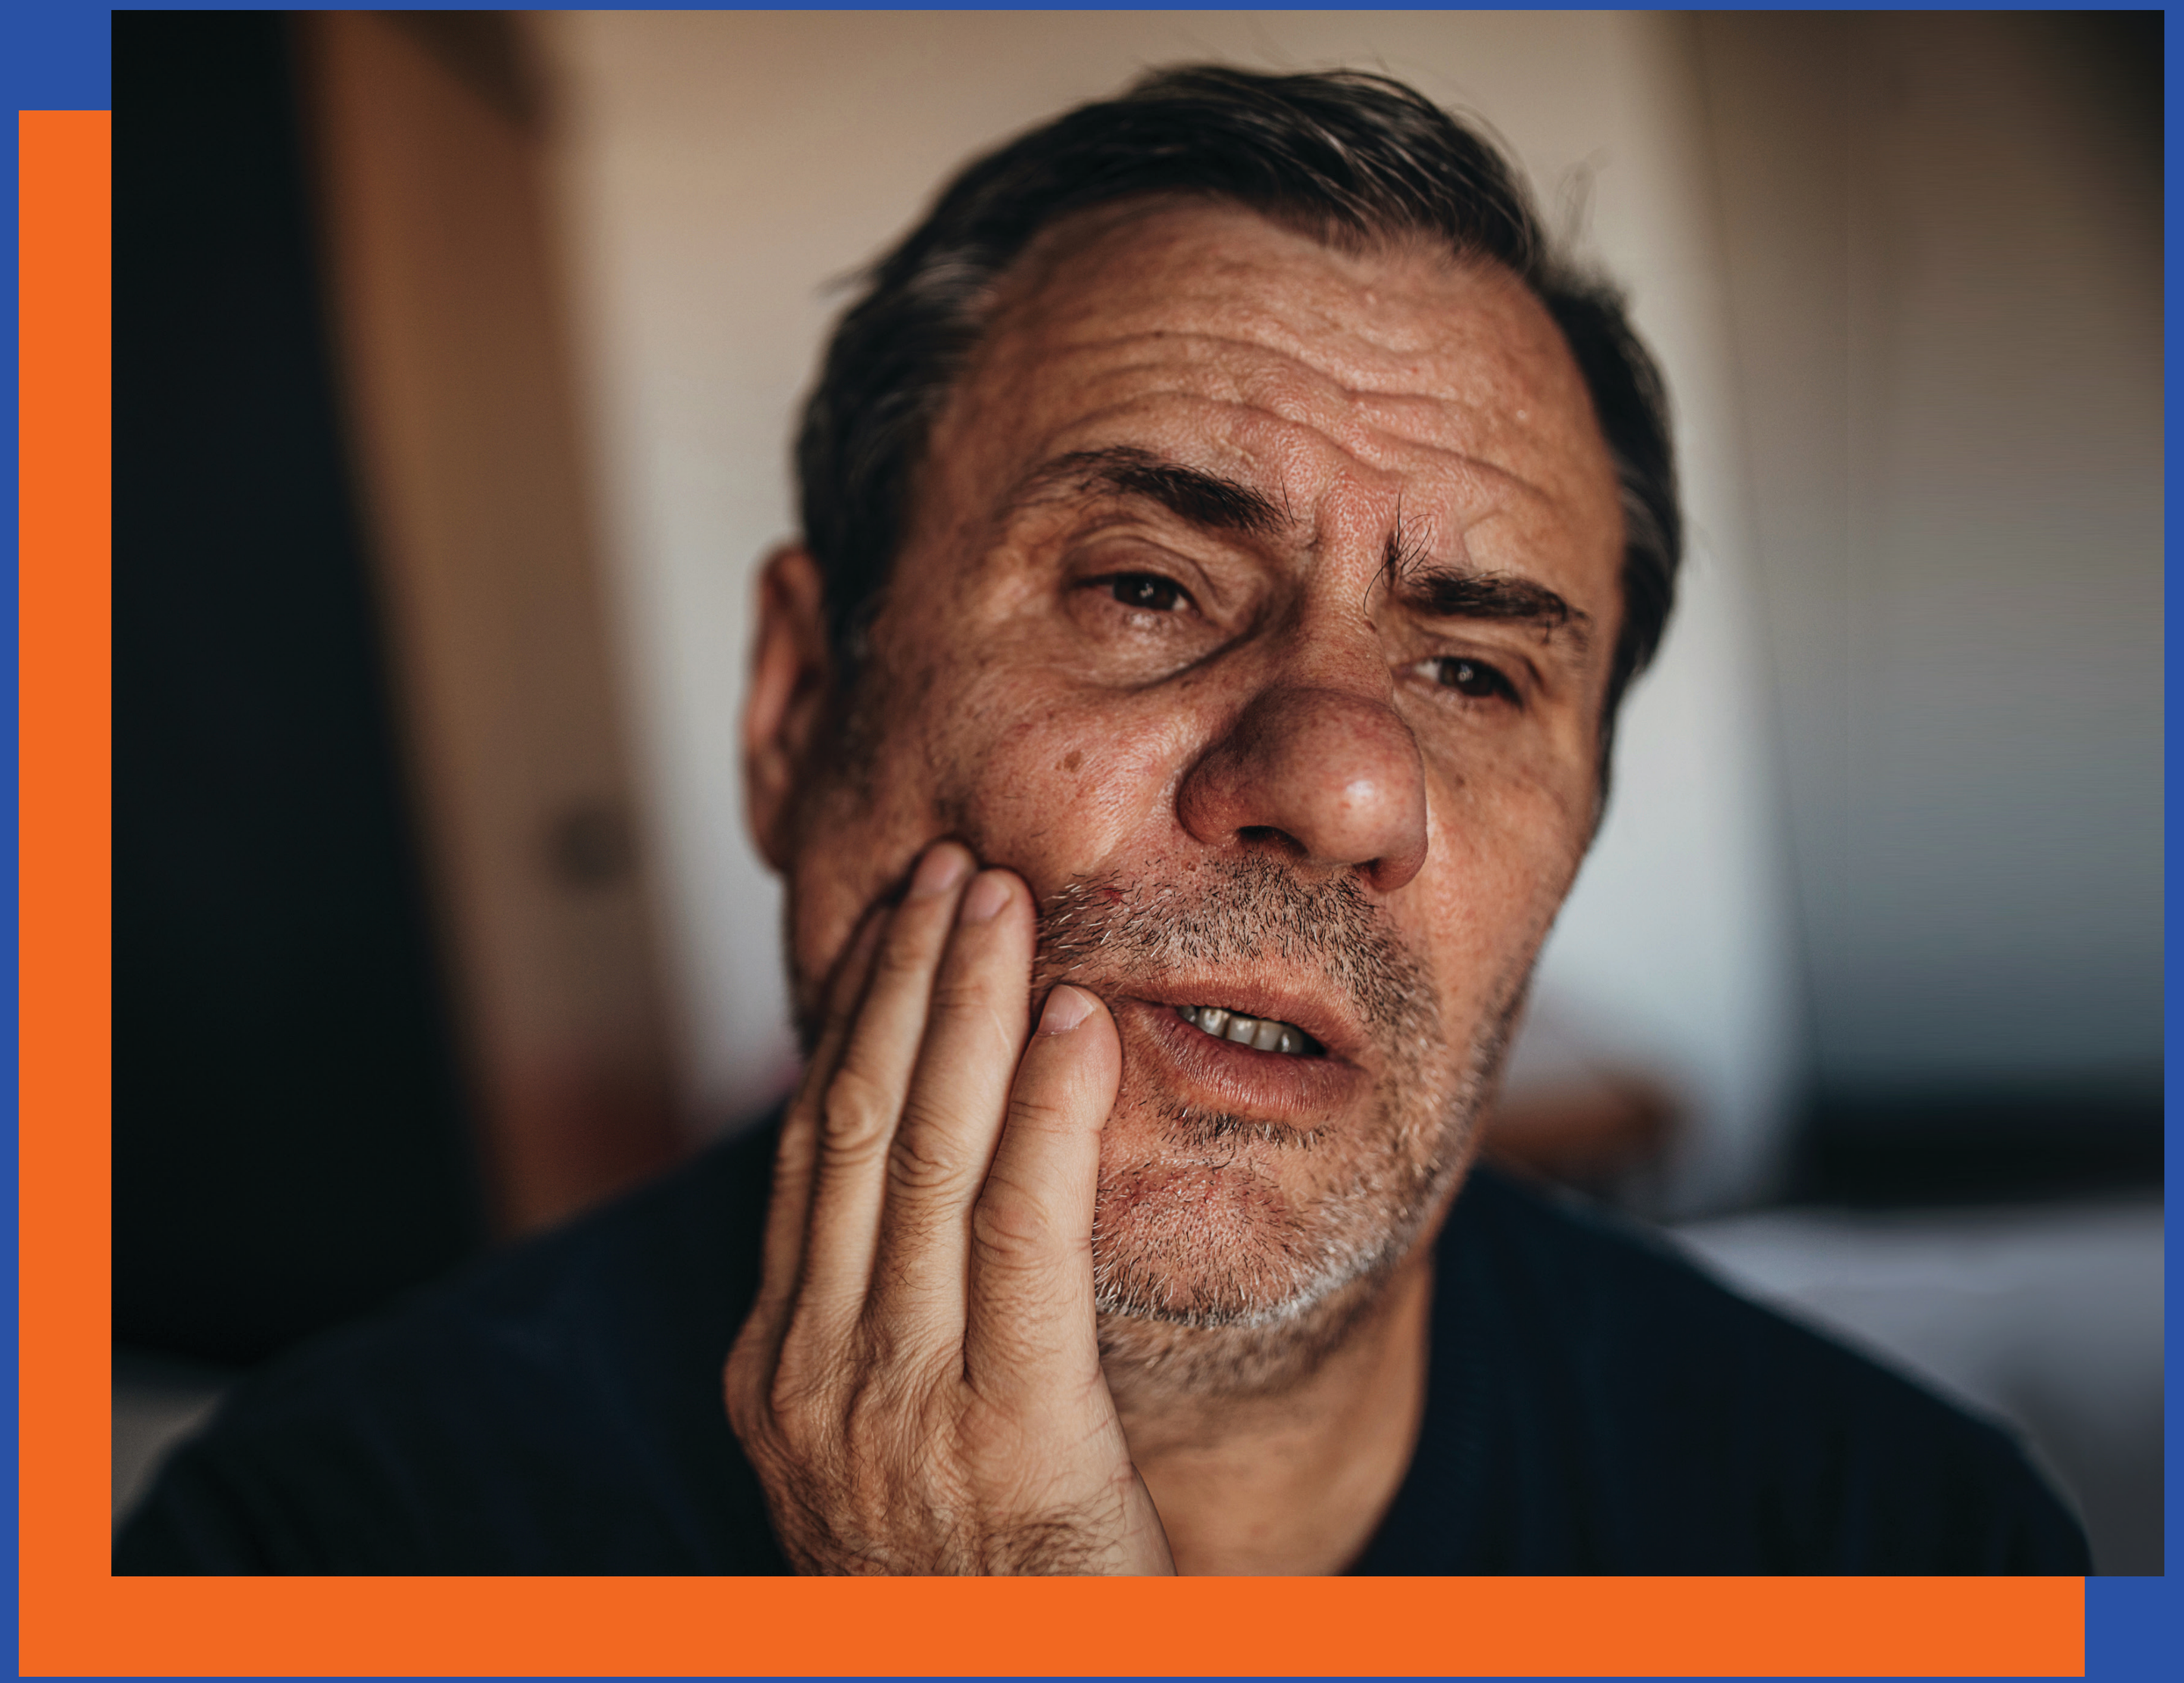

A National Survey provides insights on the use of

Home Remedies in America.

Discussion

- Study participants who reported using a home remedy to alleviate dental pain or discomfort most often reported having a toothache, swollen or bleeding gums, and/or dry mouth.
- Those who rated their overall health as fair or poor reported more often that they had oral health symptoms than those who reported overall health as good, very good, or excellent.
- Not surprisingly, there is also a correlation with annual income and the use of home remedies. The lower a participant's annual income, the more likely they were to report using a home remedy.
- The use of tobacco maybe a compounding factor with health, oral health, and the use of a home remedy for dental pain or discomfort.

Conclusion

- Patients who do not routinely seek care from an oral health provider may be more likely to turn to home remedies that may or may not be effective for managing oral pain and discomfort.
- A dental home can be an important resource when needing guidance on how to best manage dental pain or discomfort until further evaluation and follow-up care from a dental provider is possible.

Results

Oral Health Symptoms in the Last Year

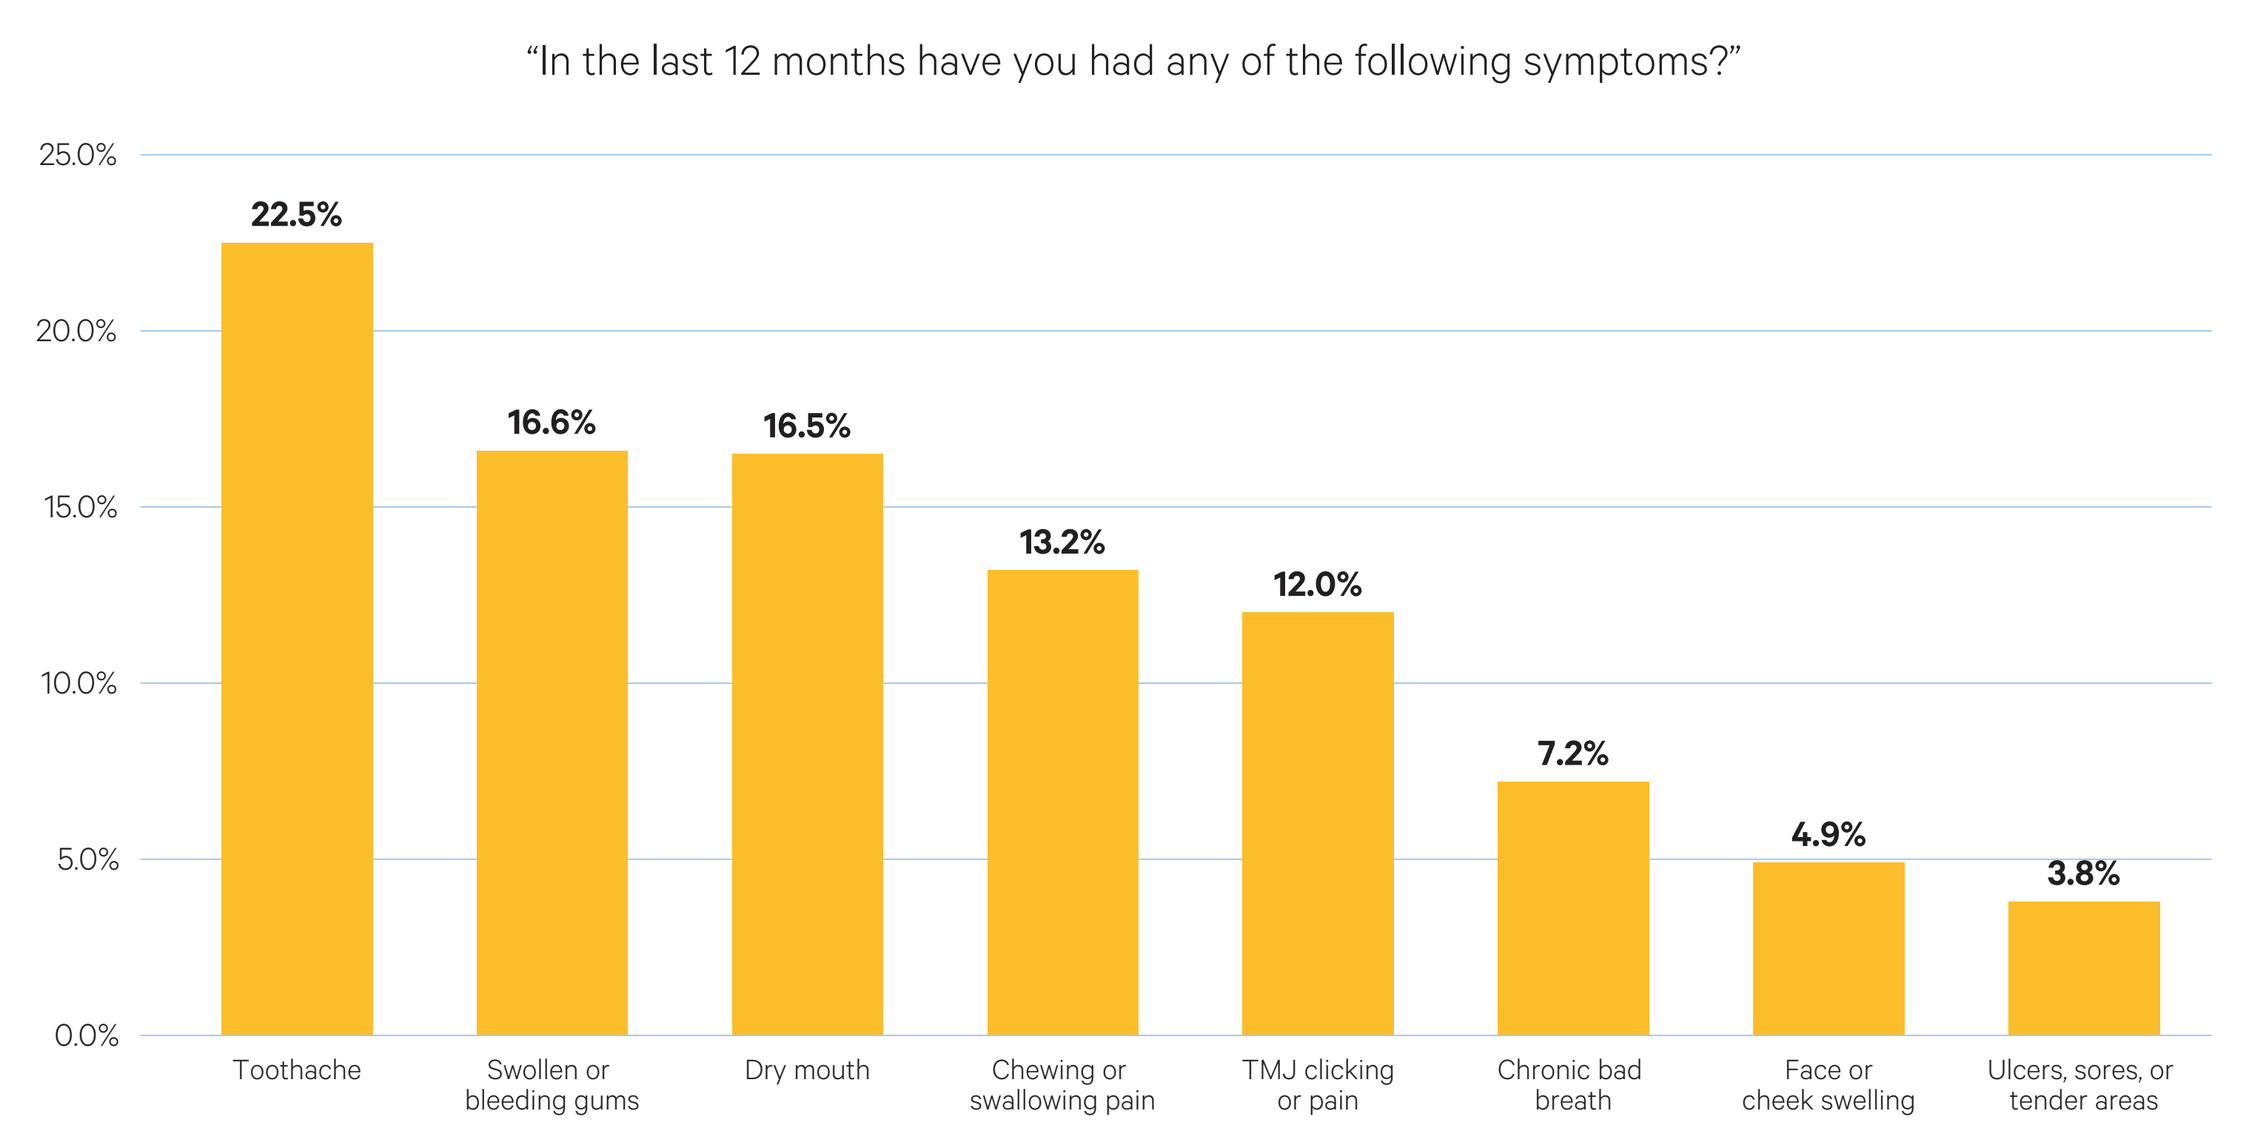
"In the last 12 months have you had any of the following symptoms?"
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
22.5%
16.6%
16.5%
13.2%
12.0%
7.2%
4.9%
3.8%
Toothache
Swollen or bleeding gums
Dry mouth
Chewing or swallowing pain
TMJ clicking or pain
Chronic bad breath
Face or cheek swelling
Ulcers, sores, or tender areas

Overall Health and Oral Health Symptoms

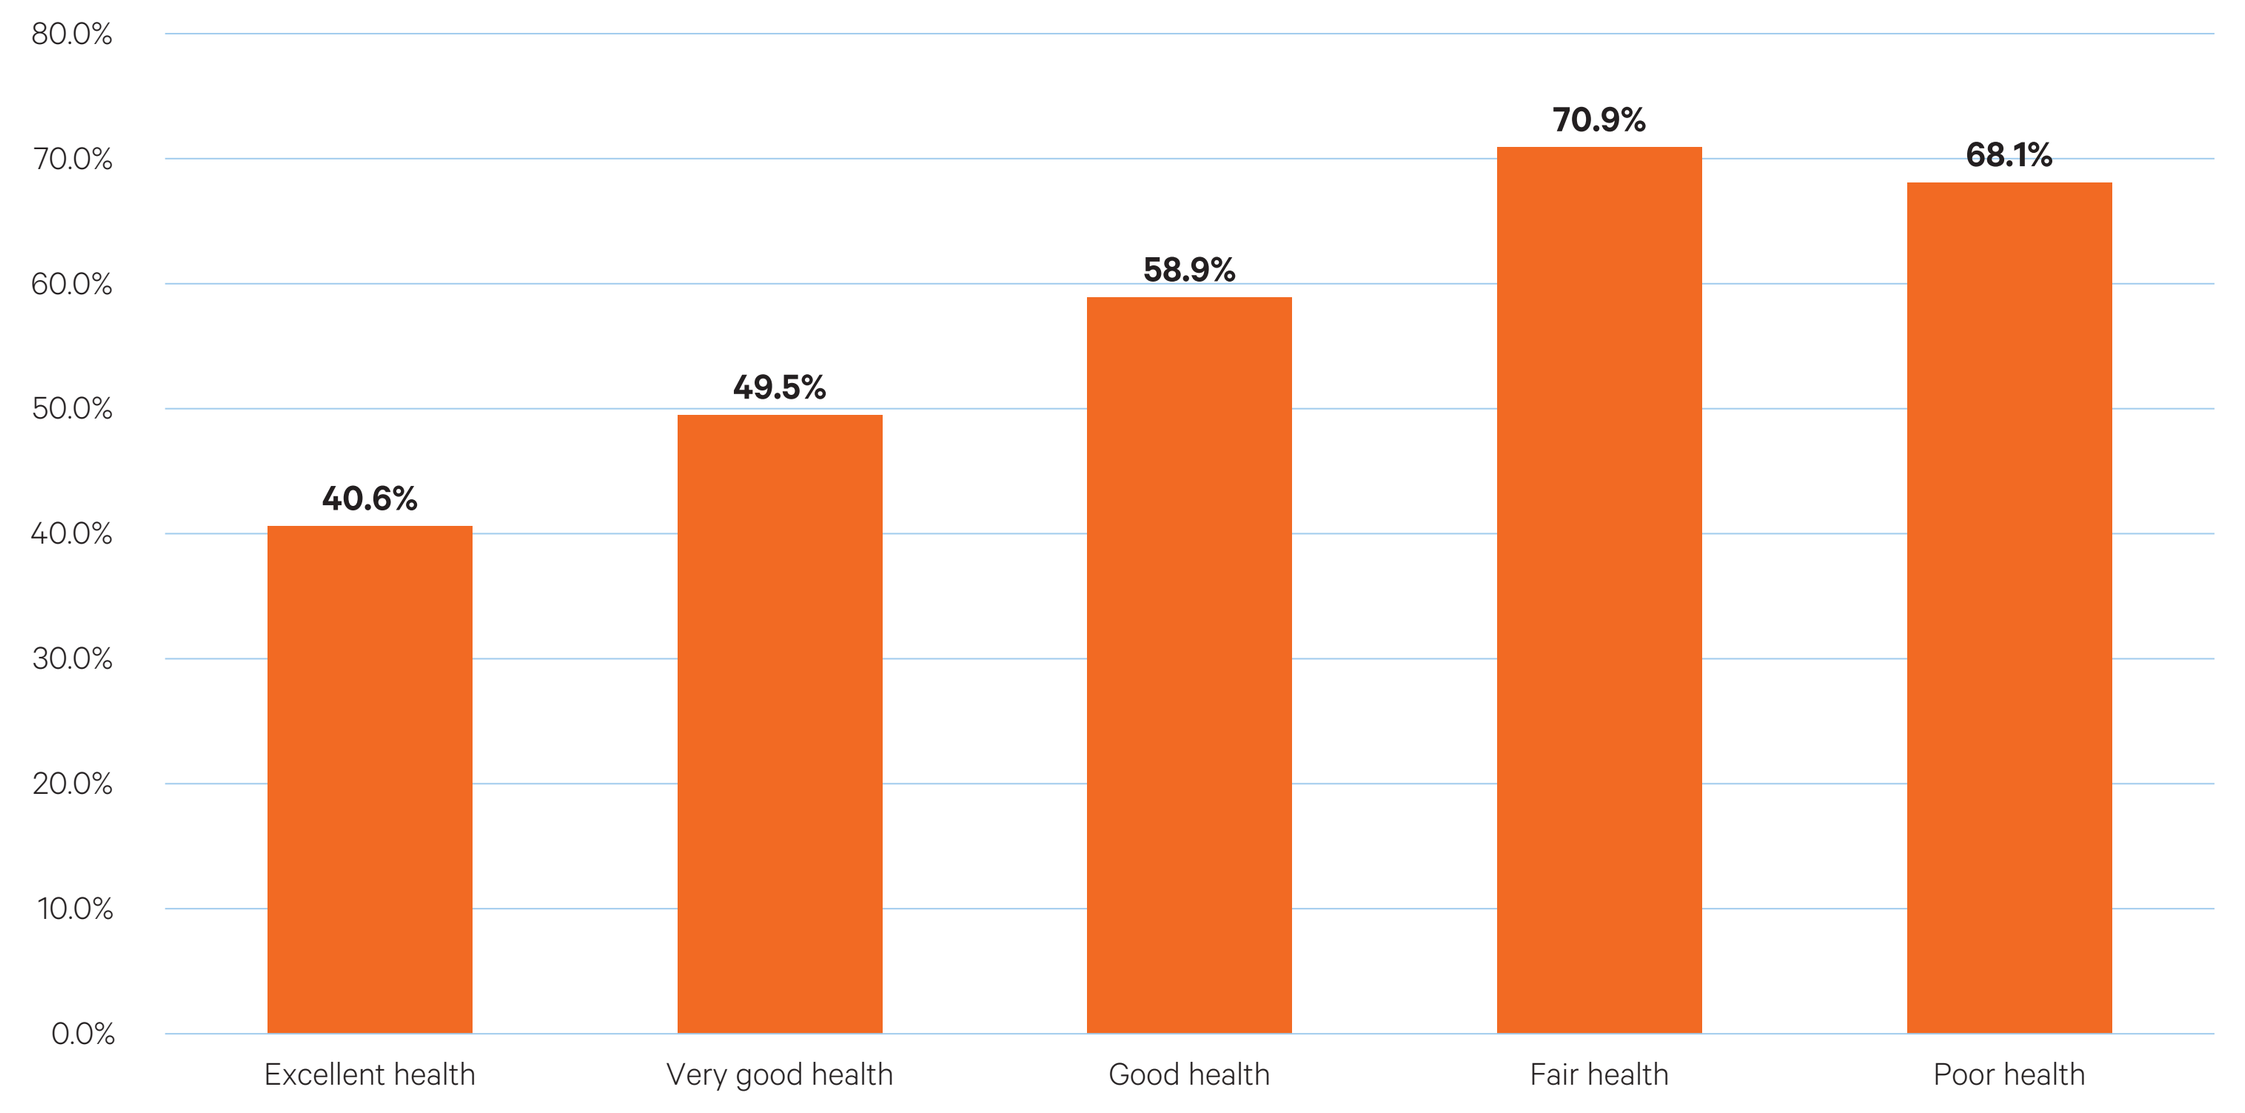
80.0%
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
40.6%
49.5%
58.9%
70.9%
68.1%
Excellent health
Very good health
Good health
Fair health
Poor health

Home Remedies Used for Dental Pain or Discomfort

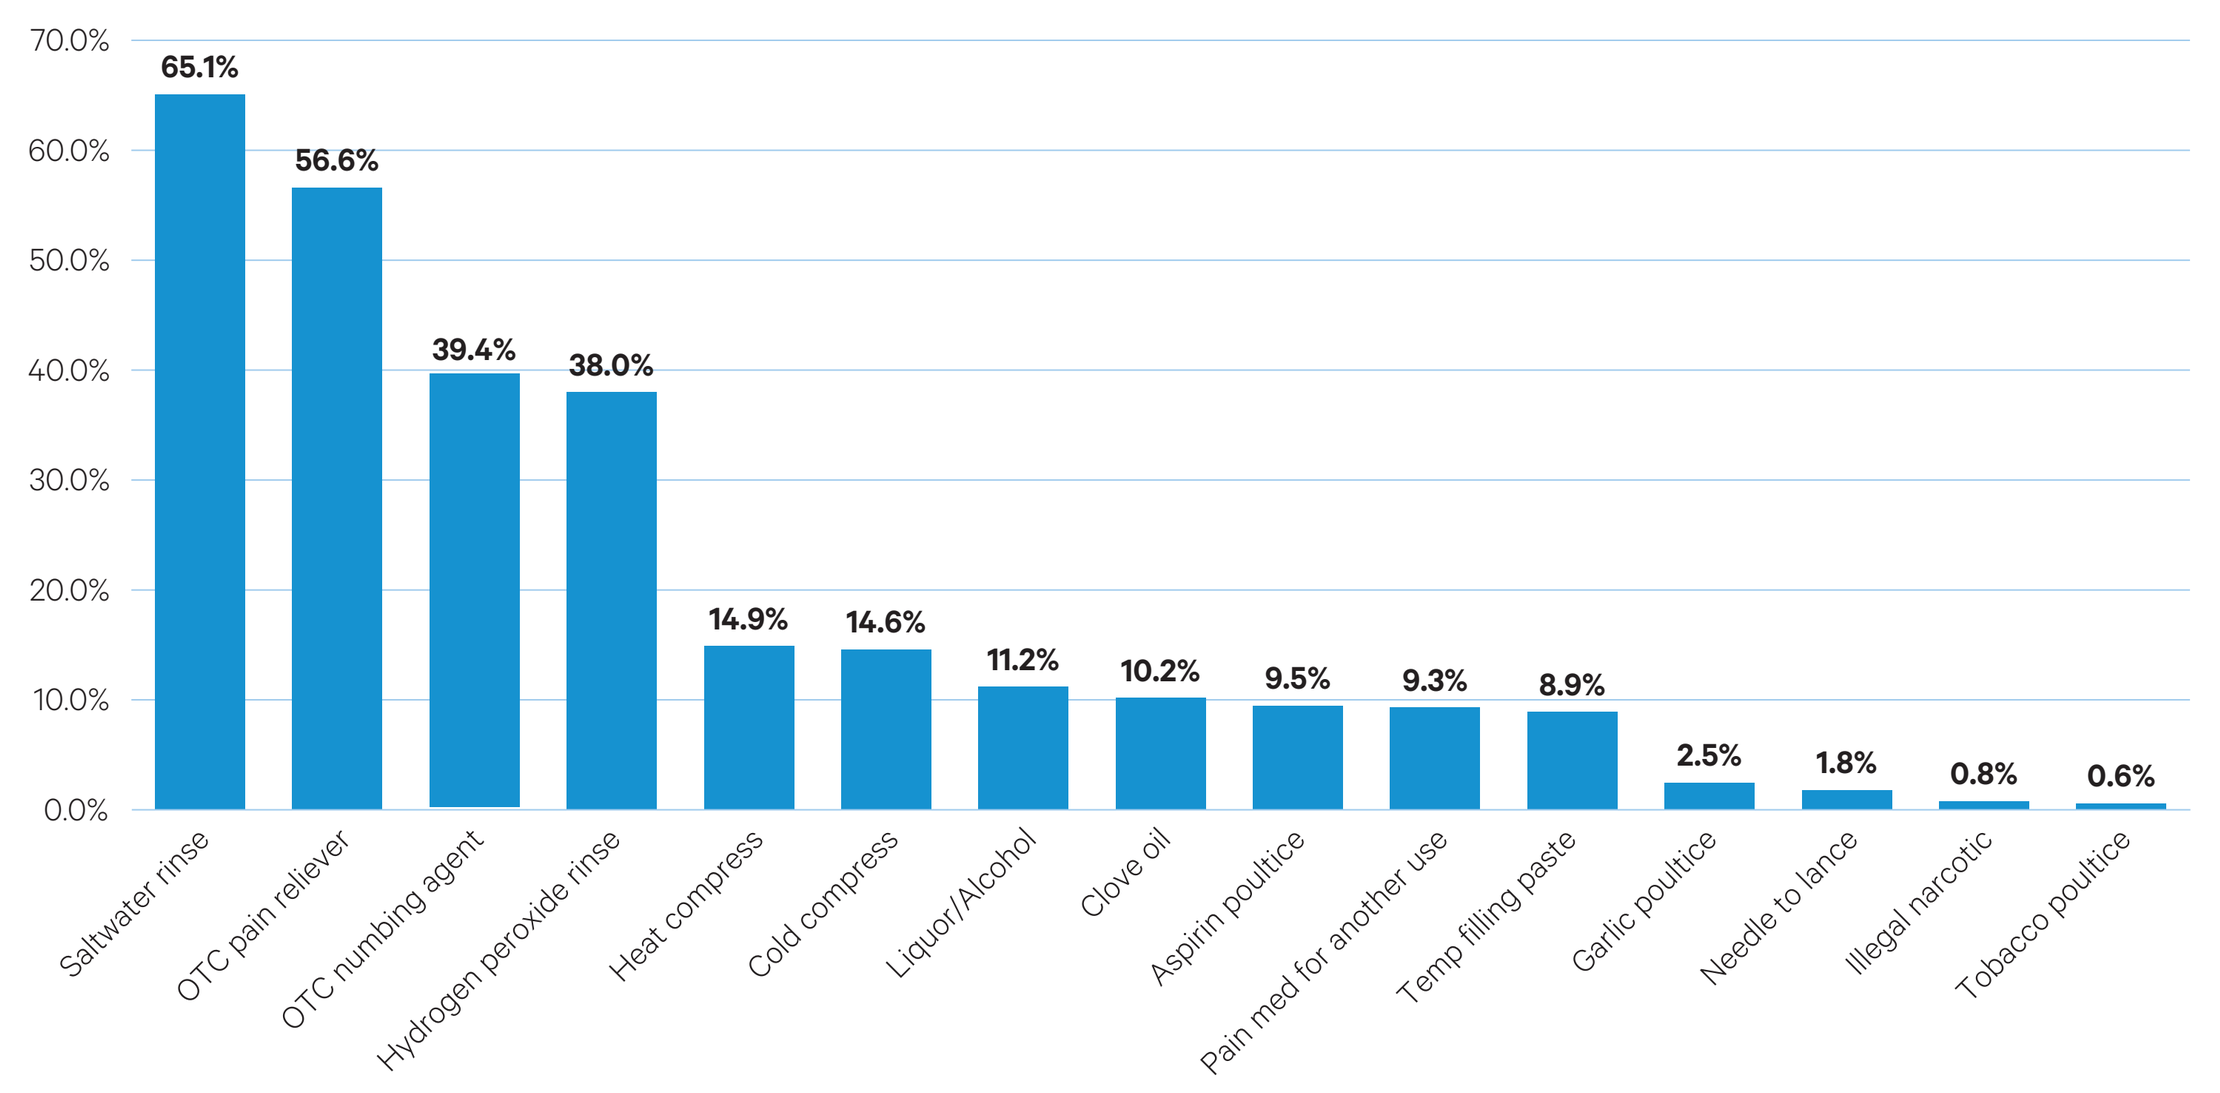
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
65.1%
56.6%
39.4%
38.0%
14.9%
14.6%
11.2%
10.2%
9.5%
9.3%
8.9%
2.5%
1.8%
0.8%
0.6%
Saltwater rinse
OTC pain reliever
OTC numbing agent
Hydrogen peroxide rinse
Heat compress
Cold compress
Liquor/Alcohol
Clove oil
Aspirin poultice
Pain med for another use
Temp filling paste
Garlic poultice
Needle to lance
Illegal narcotic
Tobacco poultice

Tobacco Use and Home Remedy Use

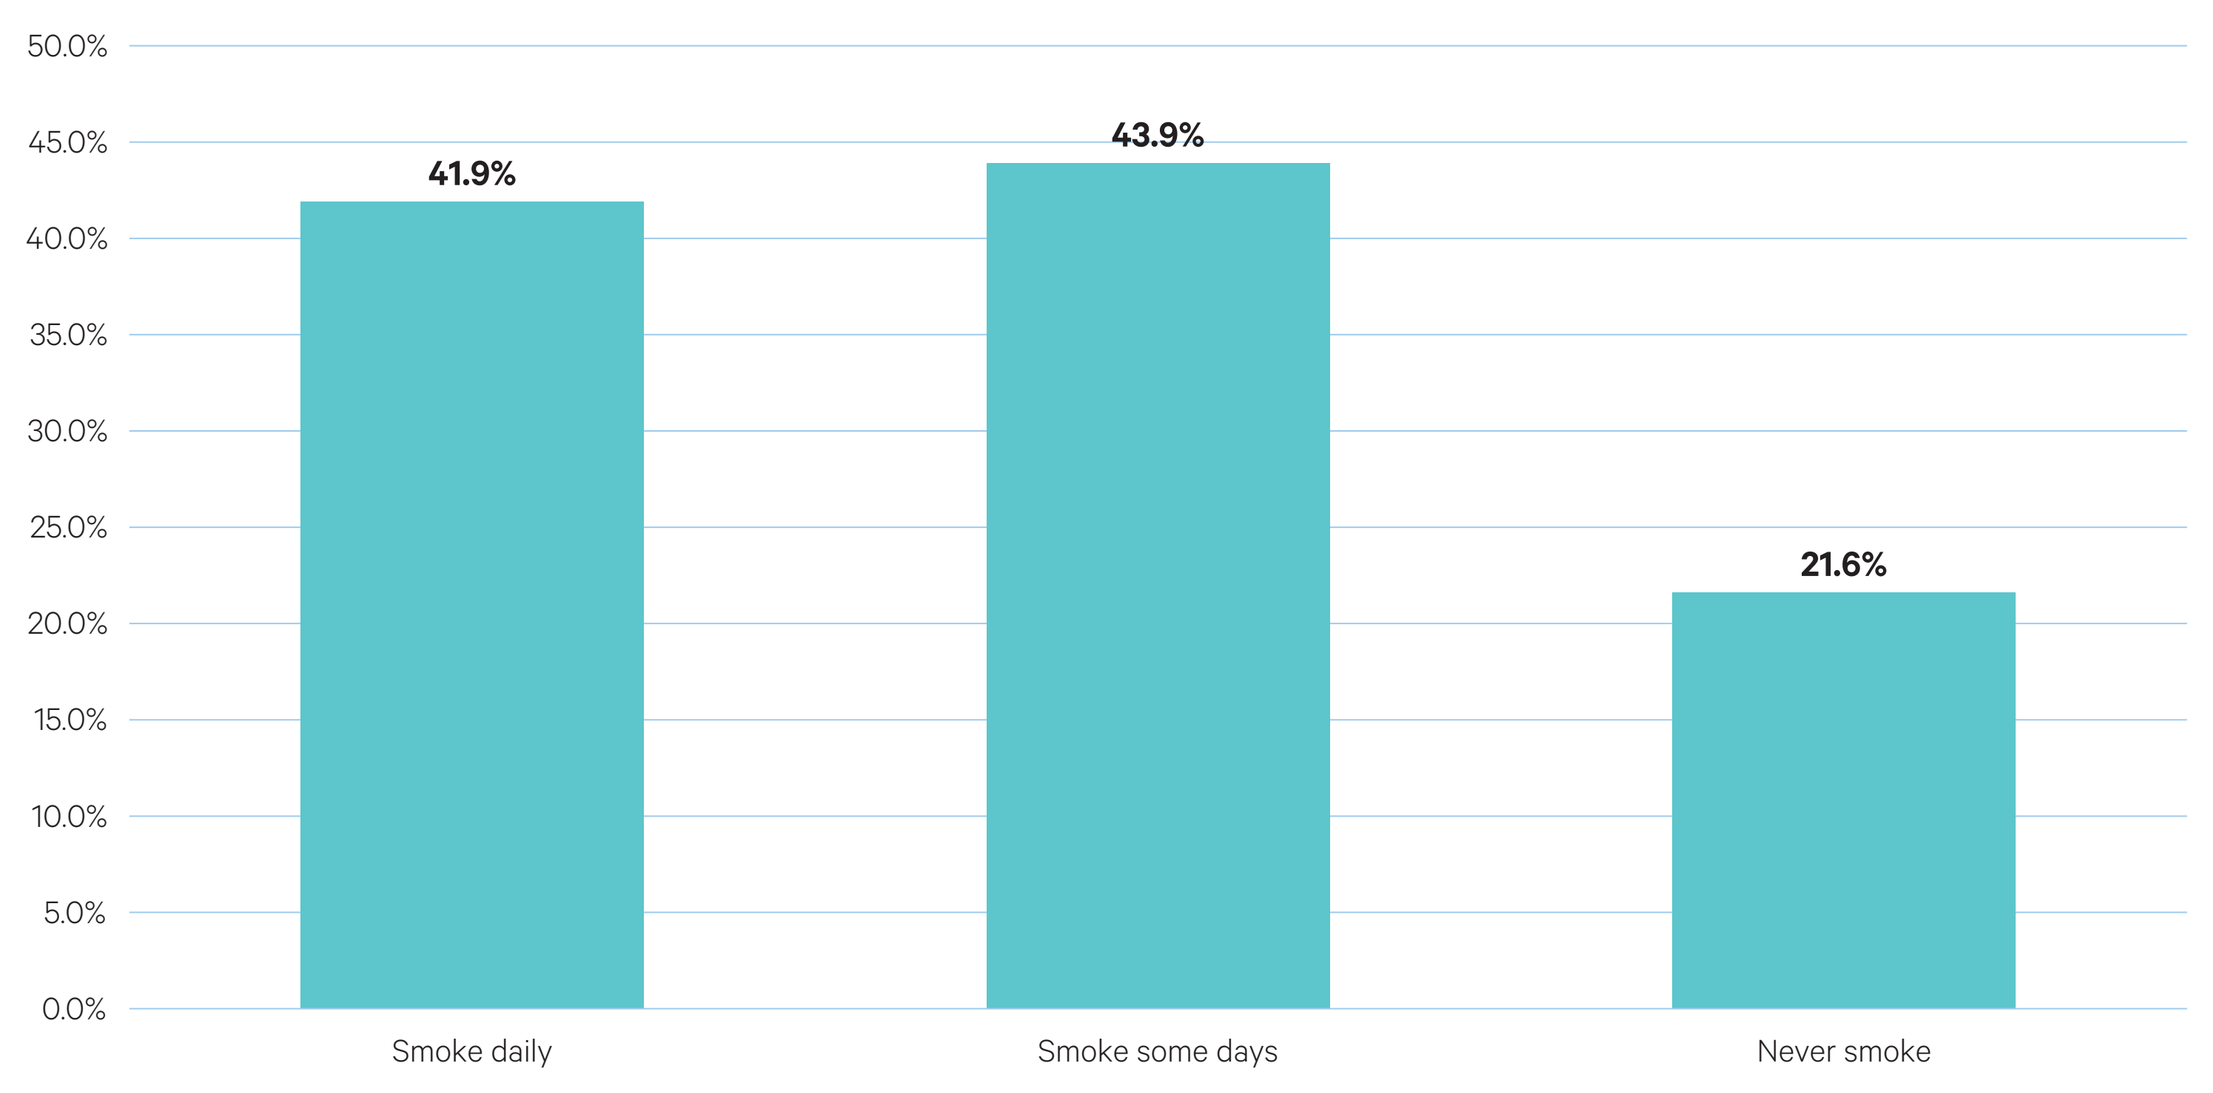
50.0%
45.0%
40.0%
35.0%
30.0%
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
41.9%
43.9%
21.6%
Smoke daily
Smoke some days
Never smoke

Home Remedy Use and Annual Income

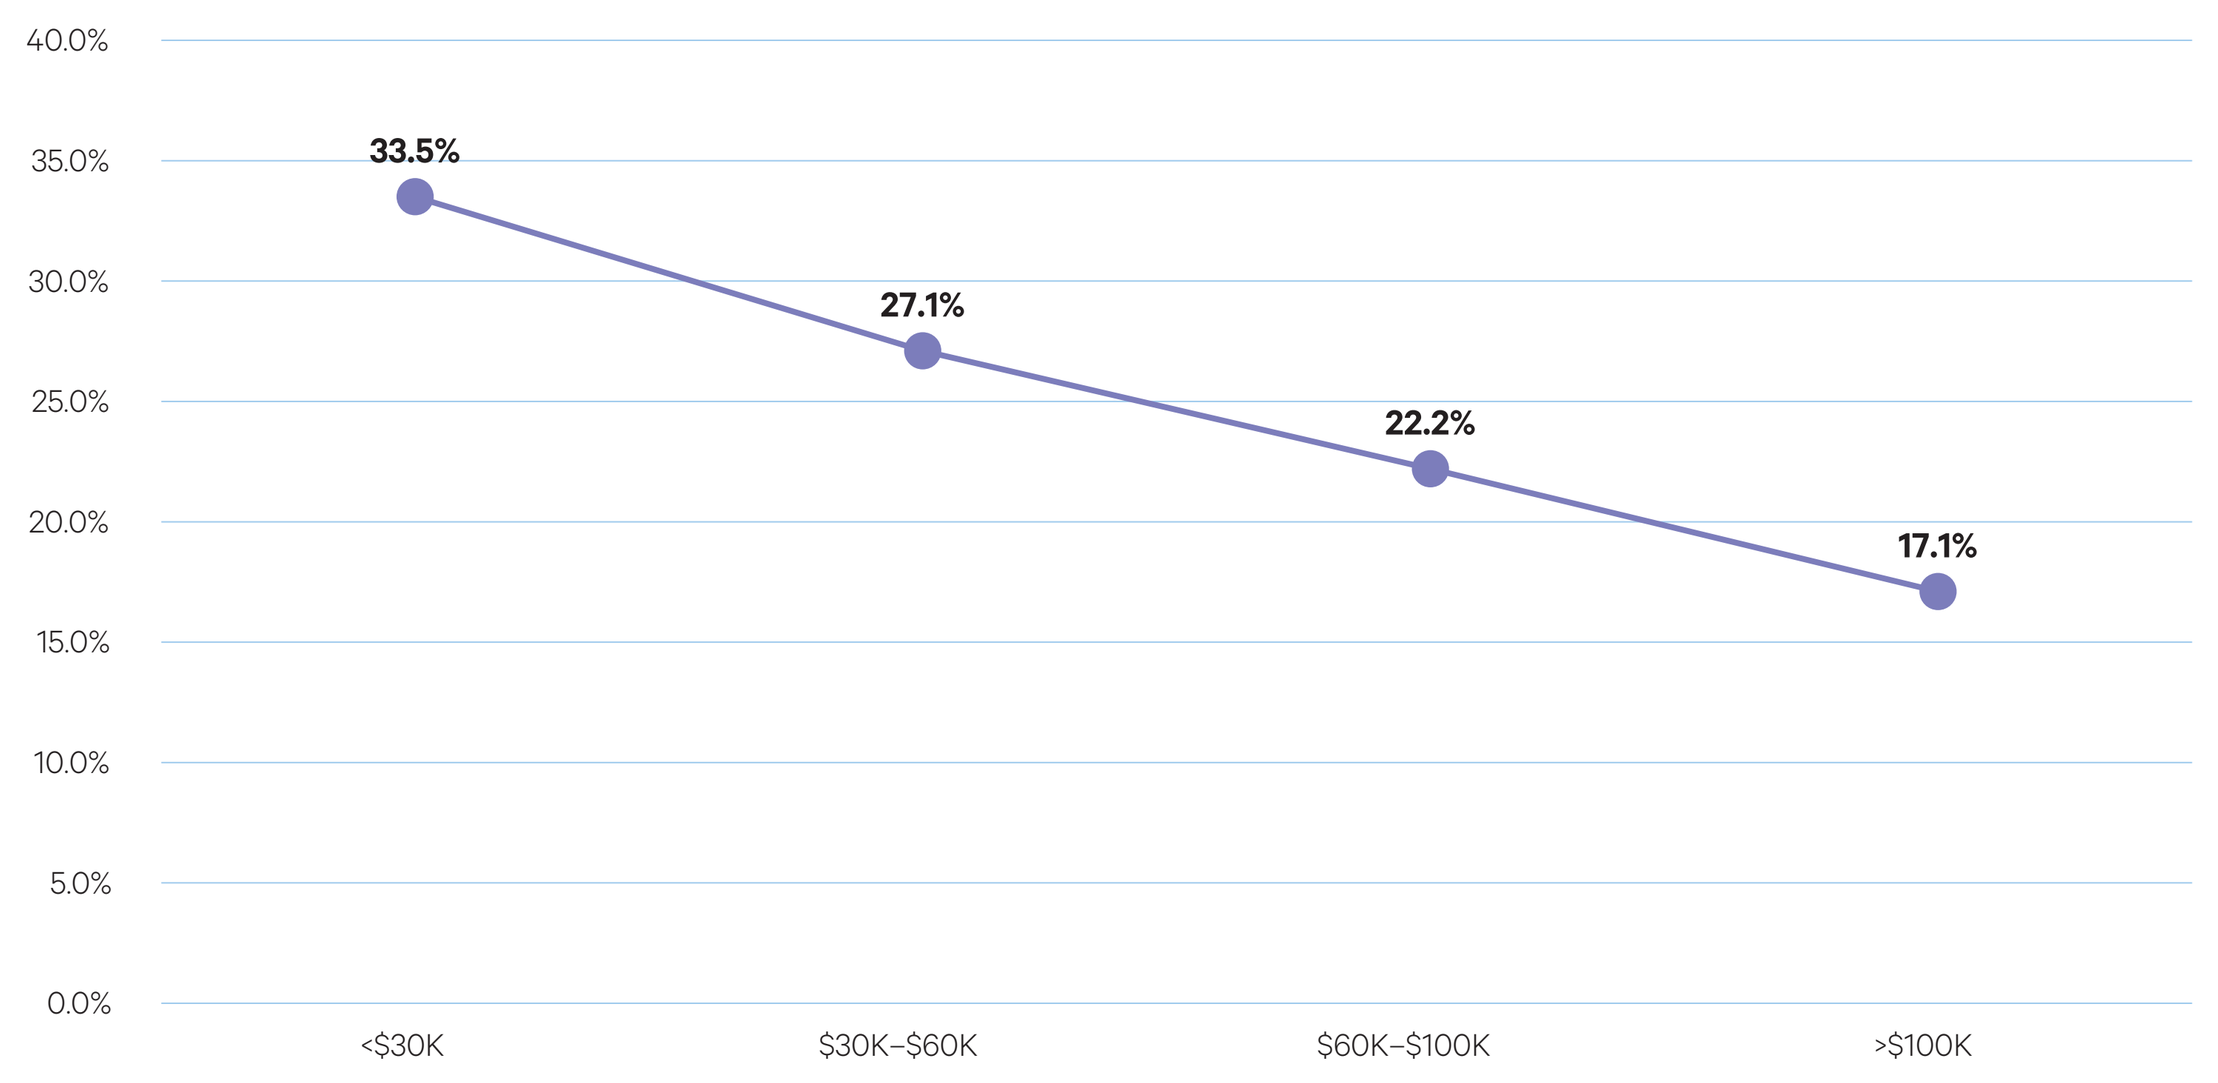
40.0%
35.0%
30.0%
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
33.5%
27.1%
22.2%
17.1%
<$30K
$30K–$60K
$60K–$100K
>$100K

CareQuest Institute for Oral Health®

Additional Resources

Heaton, Lisa J., Santoro, Morgan, Martin, Paige, and Tranby, Eric P. Cost, Race, and the Persistent Challenges in Our Oral Health System. Boston, MA; June 2023. DOI: 10.35565/CQI.2023.2005

Janakiram C, Okunev I, Tranby EP, Fontelo P, Iafolla TJ, et al. (2023) Opioids for acute and chronic pain when receiving psychiatric medications. PLOS ONE 18(9): e0286179. https://doi.org/10.1371/journal.pone.0286179