Beyond the Screen: How Dental Providers are Using Teledentistry to Meet Patient Needs

K Schroeder, RDH, MS; M Santoro, MPH; P Martin, BS; S Raskin, PhD, MPH; E Tranby, PhD

Introduction

We know little about how oral health professionals are using teledentistry in day-to-day dental practice in the aftermath of a global pandemic. Learning about teledentistry in use can help guide future work to develop standards, create best practices, and inform policy. In this study, we looked to oral health professionals who use teledentistry to learn more about the types of teledentistry being used for oral health care delivery in various scenarios.

Methods

In this **mixed-methods study**, qualitative data from provider experiences were used to help explain the quantitative data surrounding teledentistry utilization.

- **Quantitatively,** we analyzed teledentistry claims data and patient electronic health record (EHR) data to identify patterns of teledentistry service delivery.
- **Qualitatively,** we interviewed dental providers (N=13) about their teledentistry use, then analyzed interviews to identify major themes.

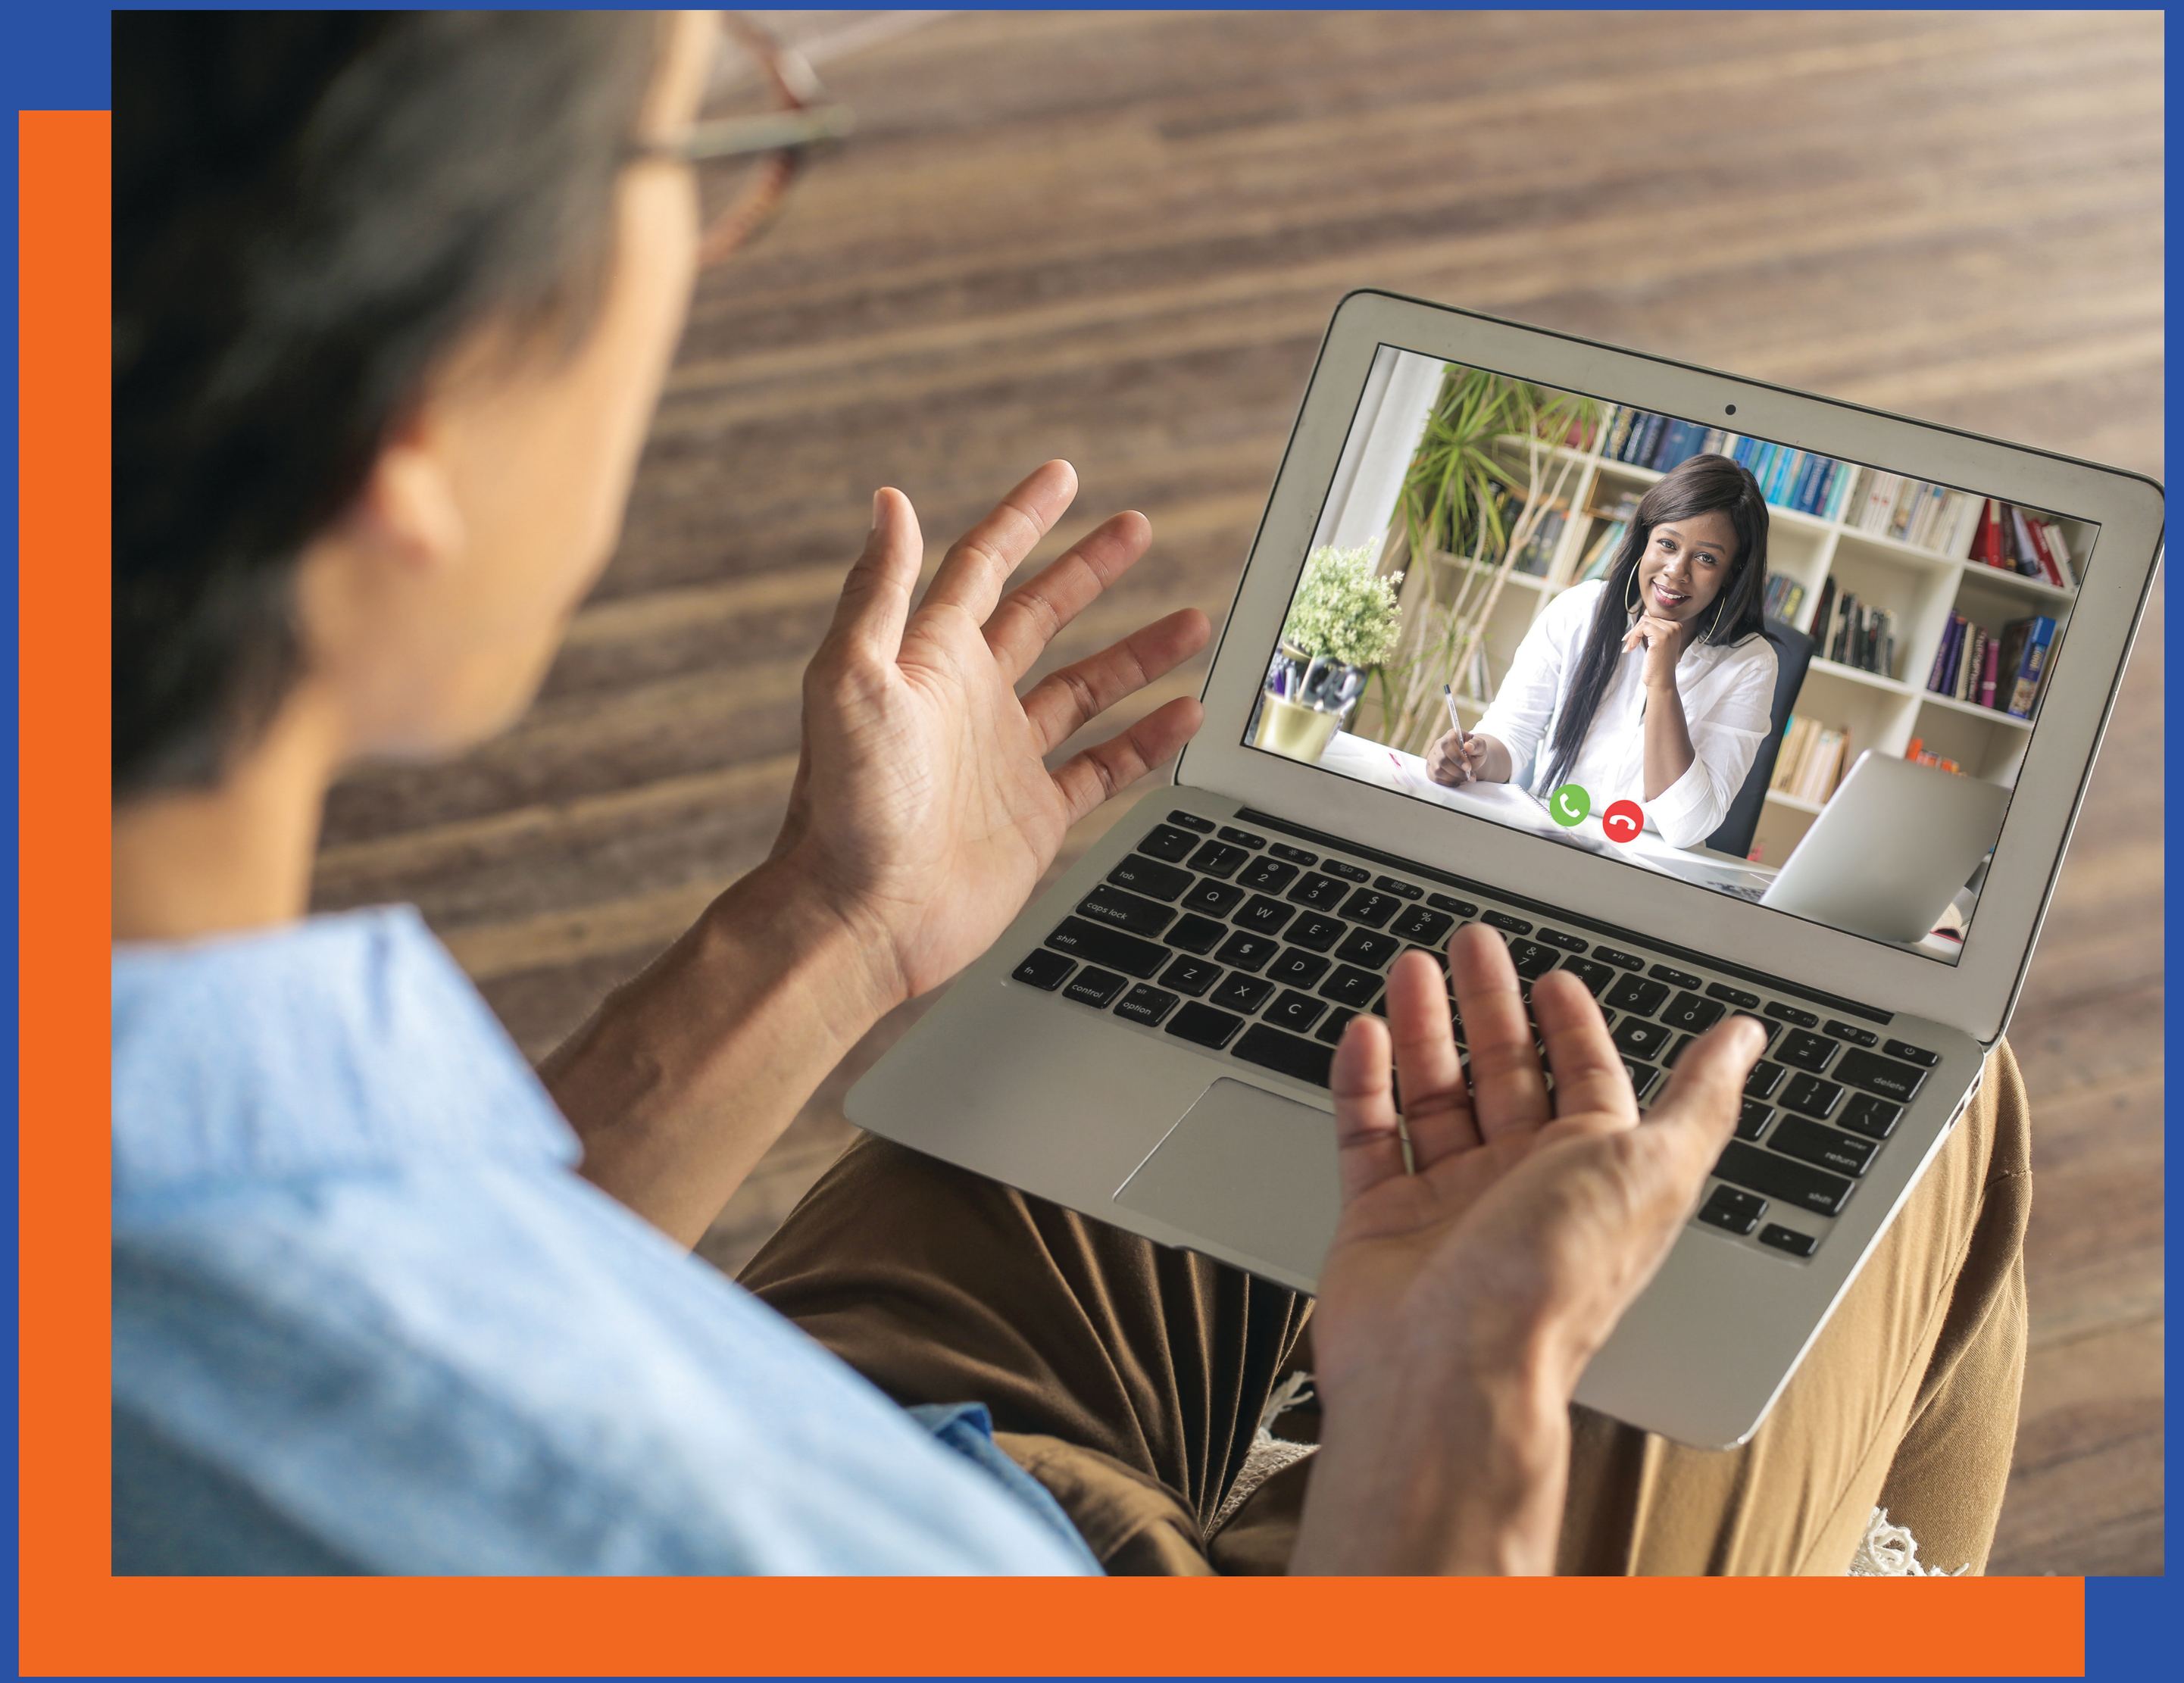

Teledentistry variations for equitable access to oral health care.

Scan for more resources from CareQuest Institute for Oral Health

Discussion

- Providers have success with using teledentistry in clinical and community settings, while also facing challenges such as variable technical skills among staff members and patients; broadband limitations among patients; and payor limitations on teledentistry reimbursement.
- Video may seem to better approximate a traditional dental visit and may be favored among providers, policymakers, and other stakeholders who have optimum access to the latest technology and digital connectivity.
- Participants in our study emphasized the importance of retaining audio-only synchronous visits as an important option for reaching patients who face broadband, technology, and other barriers to attending synchronous video visits.

Conclusion

- Synchronous video teledentistry is not accessible for all populations or all dental clinic capacities. Policy that supports a broad spectrum of mode and type of teledentistry dental delivery is imperative for teledentistry to continue as a tool to reduce barriers to oral health care and to improve patient oral health outcomes.

Results

Teledentistry by Year

	2021		2022	
	N	%	N	%
Asynchronous	5,692	15.30%	5,556	13.40%
Synchronous Audio	25,011	67.10%	31,412	75.70%
Synchronous Video	6,557	17.60%	4,528	10.90%
Total	**37,260**	**100%**	**41,496**	**100%**

Subject Dental Clinic Teledentistry Utilization by Year

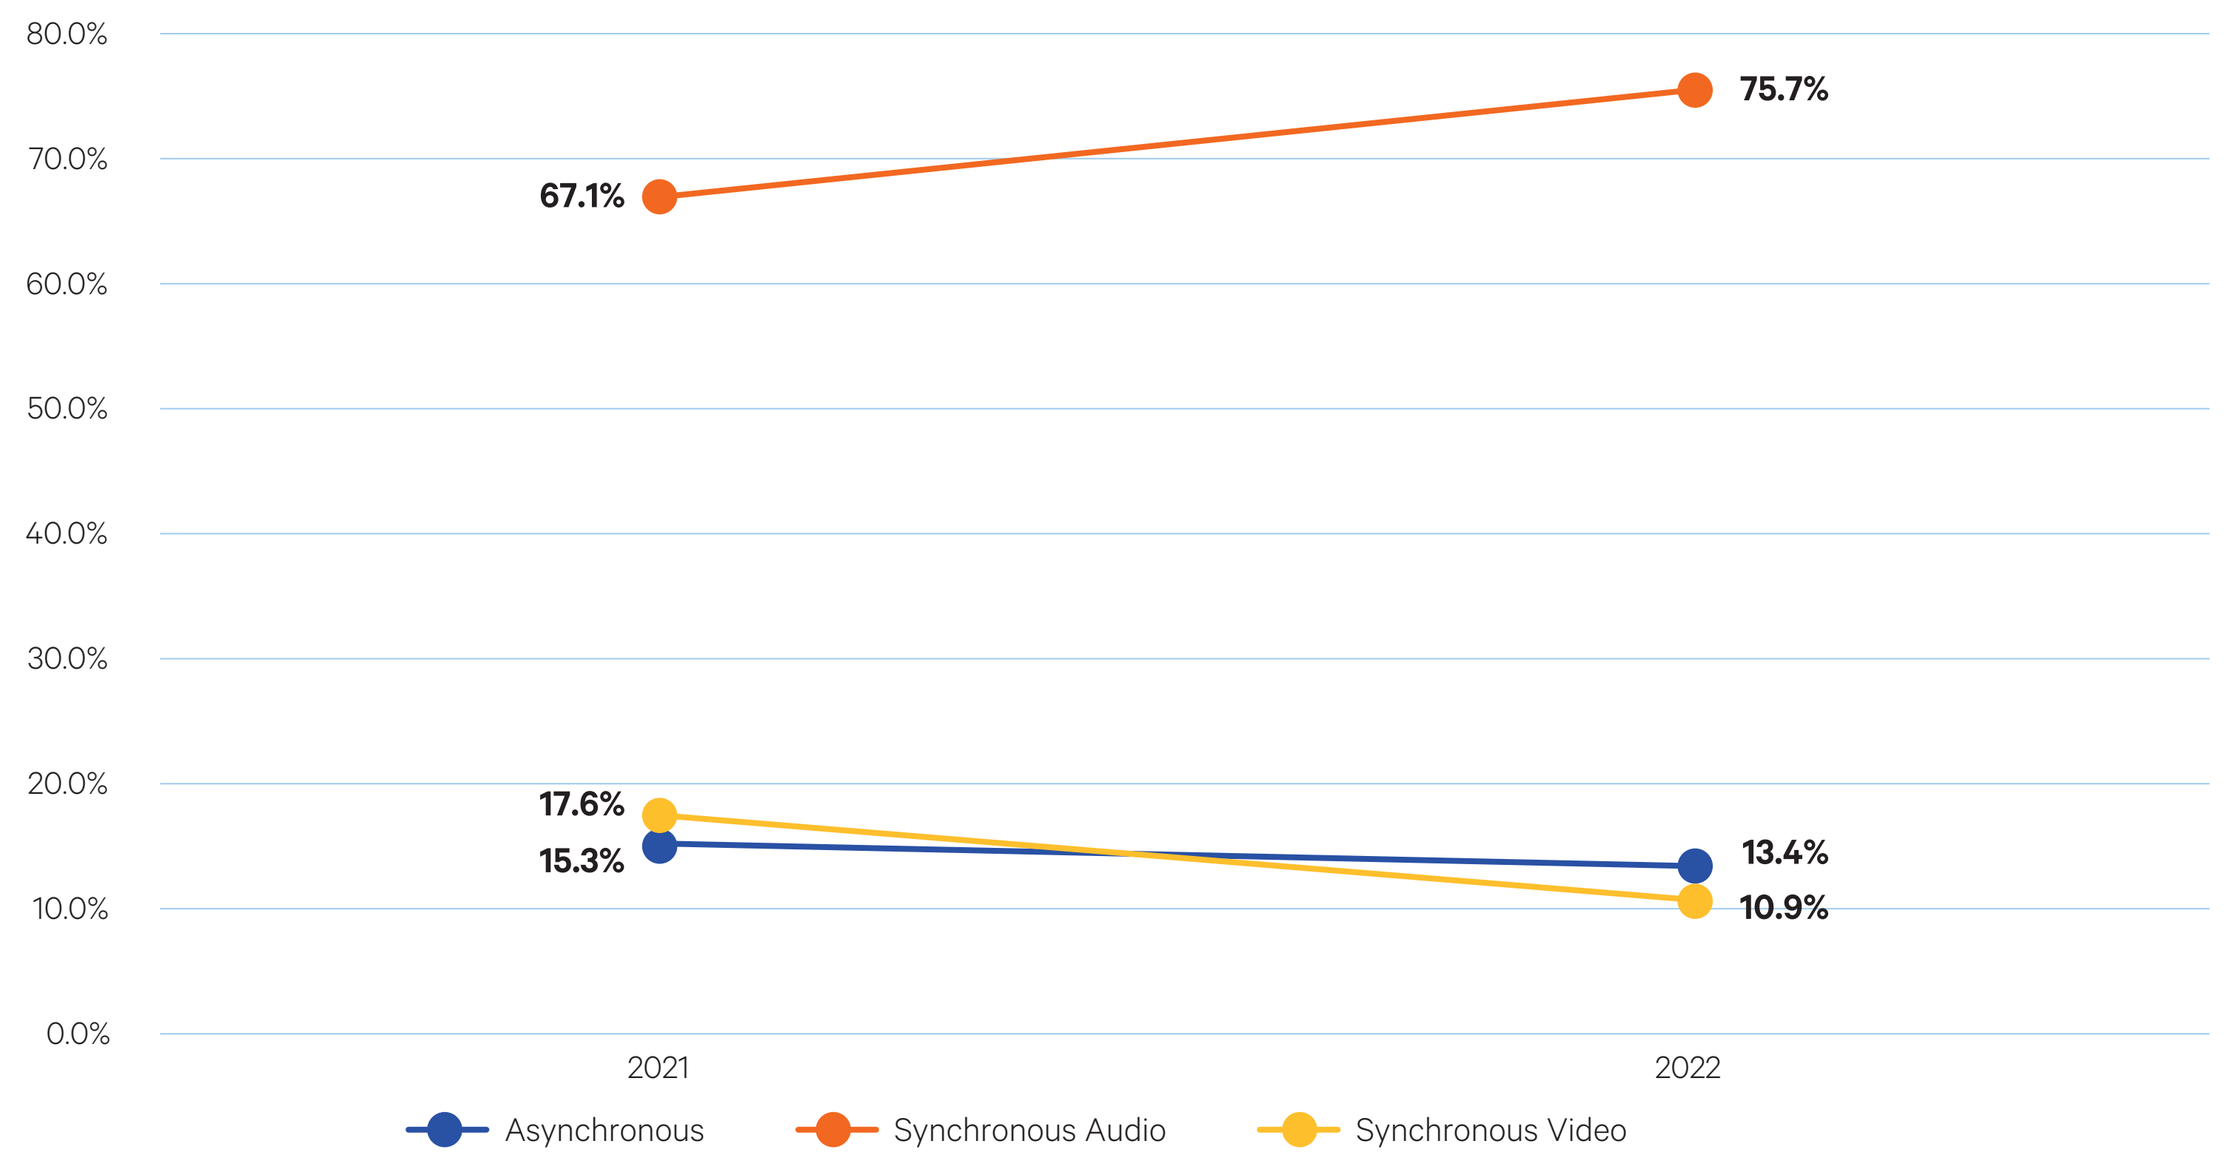
80.0%
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
67.1%
75.7%
17.6%
15.3%
13.4%
10.9%
2021
2022
Asynchronous
Synchronous Audio
Synchronous Video

Theme 1: Providers deliver oral health care services using a variety of teledentistry types.

Synchronous-Clinic Setting
"he'll (the dentist) reach out when he has time and here is when we can try to expect it… It's really smooth. Patients really love it."
Asynchronous-Clinic Setting
"…dental hygienists see patients sometimes in the medical clinic or in a health department… they're capturing those images…then at a later date, the teledentist is… making that treatment plan.
Synchronous-Community Setting
"… teledentistry that I do are ECCP (Early Childhood Caries Prevention) calls… over the phone conversation.
"… the parents do the oral examination and so I'm able to help them in different ways."

Theme 2: Teledentistry implementation involves both successes and challenges

	Successes — Clinical
Access to timely care	"A lot of times when patients call in and they're in pain and they have swelling, and they're scared. … the doctor can give them the medicine that they need to start dealing with the infection or whatever they need."
	Successes — Community outreach
Patient education	"My biggest successes are from over the phone. …we live in an area that has a really, really high decay rate for one, and also really high migrant population…"
	Challenges — Clinic and Community Outreach
Technology	"And then I would say about half the time the quality is just not there. Typically it is on the patient side…they have bad service…and that is why we are doing teledentistry in the first place because they live rurally."
Payor	"…my only concern along the way is how is it going to be covered…. is there this unnecessary extra stress around the financials of it?"

Theme 3: Variation and flexibility are an essential for optimizing teledentistry.

"I really truly hope that we're able to continue just to be over the phone because I think that there's a lot of access issues if it is something where it's only video."
"Other factors… (is they) end up losing service so trying to be in a place with service because we do send them like text messages for the consent form."
"… sometimes the teledentistry platform works and if it doesn't, I will utilize a phone and photos that they can send to our email address because I'd rather help them and have it not be officially through the teledentistry platform than say, you know, tough luck and see you in a month
"Sometimes the internet doesn't help… you lose connection so have to dial back again to the patient, hopefully they pick up."
"…[patients] more advanced in age and feel like they might struggle with the technology side of it (teledentistry) which is a fair concern and that's why we don't push it… it not like a requirement."

CareQuest Institute for Oral Health®

Additional Resources

CareQuest Institute for Oral Health. *Teledentistry: What to Know and How It Works.* Boston, MA: CareQuest Institute for Oral Health, 2021. Accessed October, 2023. https://www.carequest.org/system/files/CareQuest-Institute-Teledentistry-What-To-Know.pdf

Howell, Scott E and Fukuoka, Brooke. "Teledentistry for Patient-centered Screening and Assessment". *Dent Clin North Am* 66, 2(April 2022): 195-208. doi: 10.1016/j.cden.2022.01.002

Tiwari, Tamanna; Diep, Vuong; Tranby, Eric; Thakkar-Samtani, Madhuli; and Frantsve-Hawley, Julie. "Dentist perceptions about the value of teledentistry." *BMC Oral Health* 22,1: 176. (May 2022). doi: 10.1186/s12903-022-02208-z.

Tranby, Eric and Thakkar Samtani, Madhuli. *Teledentistry is an Effective Tool to Triage Patients and Save Money.* Boston, MA: CareQuest Institute for Oral Health, 2020. DOI: 10.35565/CQI.2020.2026. Accessed October, 2023.